^{111}In-labeled Nimotuzumab Modified with Nuclear Localization Sequences (NLS) for Targeting Herceptin® Resistant Breast Cancer Cells

Aisha Fasih[1] and Raymond M. Reilly[1,2,3]

[1]Department of Pharmaceutical Sciences, University of Toronto, [2]Toronto General Research Institute, [3]University Health Network

Abstract

Objective: Nimotuzumab (Nmab) is a monoclonal antibody selectively targeting cancer cells overexpressing epidermal growth factor receptor (EGFR). Abnormal up-regulation of EGFR is found to be present in breast cancer (BC) cells which are resistant to trastuzumab (Herceptin®) and is associated with poor prognosis. Currently, there are few agents that target EGFR on BC cells with acquired resistance to trastuzumab. Thus, our objective is to modify an already established anti-EGFR antibody (Nmab) with an Auger emitting radioisotope Indium-111 to target cells that are either intrinsically resistant or acquire resistance to Herceptin. **Methods:** Nmab was derivatized with sulfosuccinimidyl-4-(N-maleimidomethyl) cyclohexane-1-carboxylate (sulfo-SMCC) for reaction with NLS-peptides and labeled with Indium-111 using benzyl-diethylenetriaminepentaacetic acid (bn-DTPA). The immunoreactivity of ^{111}In-NLS-Nmab was determined by its ability to displace the binding of Nmab in MDA-MB-468 human breast cancer cells. Cellular uptake and nuclear localization were evaluated in MDA-MB-468 and MCF-7 cells, which express high and very low levels of EGFR, respectively, by cell fractionation. Finally, the cytotoxicity of ^{111}In-Nmab and ^{111}In-NLS-Nmab was studied by clonogenic assays. **Results:** The characterization of ^{111}In-NLS-Nmab by instant thin layer chromatography (ITLC) and SDS-PAGE showed there were 2.09$\pm$2.21 bn-DTPA and 8.5$\pm$0.20 NLS per antibody. Furthermore, the radiochemical purity and specific activity was found to be 99.23$\pm$0.12% and 350$\pm$9.50 MBq/mg respectively. The dissociation constant for binding of ^{111}In-NLS-Nmab to MDA-MB-468 cells was 9.8$\pm$0.86 nM. In the case of ^{111}In-NLS-Nmab 31.2$\pm$2.59% of the radioactivity associated with MDA-MB-468 cells was found in the nucleus compared to only 16.0$\pm$0.63% for ^{111}In-Nmab. For comparison, ^{111}In-EGF had only 2.1$\pm$0.11% internalized in the nucleus. Moreover, the internalization was significantly lower in MCF-7 cells which express low levels of EGFR similar to that on normal epithelial tissues. The clonogenic survival of MDA-MB-468 cells exposed for 3 days to 20nM of Nmab, NLS-Nmab, ^{111}In-Nmab and ^{111}In-NLS-Nmab was 60$\pm$7.04%, 61$\pm$6.5%, 21$\pm$5.3%, and 2.8$\pm$0.16% respectively. The CS of MCF-7 cells exposed to ^{111}In-NLS-Nmab (40 nM) was 82.6±6.4% vs. 80.0±1.6% for ^{111}In-Nmab ($p>0.05$) and 95±19.4% for Nmab ($p>0.05$). Preservation of selectivity for abnormal EGFR overexpression of the Nmab conjugate supports further development of this potent conjugate despite ubiquitous EGFR expression in normal tissues.

Background

In the past our laboratory has developed a novel radiotherapeutic agent, ^{111}In-labeled human EGF (^{111}In-hEGF) which exploits the overexpression of EGFR found in 30-50% of cases of ER-negative, hormone resistant and poor prognosis breast cancers (BC)[1, 2]. Although ^{111}In-hEGF exhibited potent cytotoxic effects towards EGFR-overexpressing MDA-MB-468 cells alone/or in combination with other drugs such as gefitinib, it had a very short half-life (24-36 mins) in mice and was also eliminated very rapidly from the blood after intravenous injection in BC patients. This rapid elimination resulted in low tumour uptake. As an alternative, in this study we now examine the EGFR-dependent cytotoxicity of the humanized IgG_1 mAb Nmab (h-R3) (YM-BioSciences) labeled with ^{111}In which is expected to have a longer half-life and greater tumour uptake.

Nmab binds to the extracellular domain of EGFR and blocks ligand activation, resulting in inhibition of EGFR activation and cancer cell proliferation as well as production of VEGF. Severe side-effects are rarely observed with Nmab, in contrast to the currently approved high-affinity anti-EGFR antibodies, cetuximab (Erbitux®) and panitumumab (Vectibix®). The severe side-effects with the other antibodies are predominantly caused by binding to EGFR in normal tissues. Nmab's improved safety profile is a result of selectively targeting cancer cells overexpressing EGFR due to intermediate affinity. The selective targeting of cancer cells makes Nmab the preferred anti-EGFR antibody for conjugation.

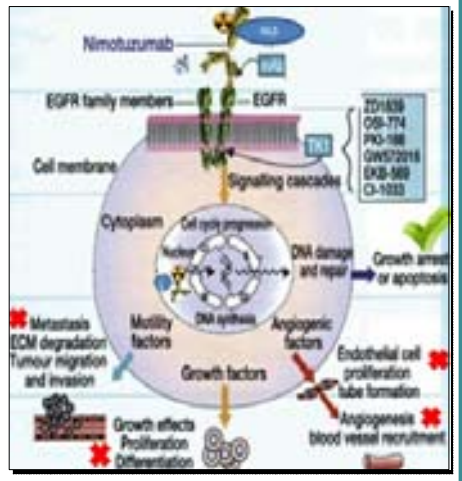
Nimotuzumab
EGFR family members
EGFR
Cell membrane
Signaling cascades
Cytoplasm
Cell cycle progression
Nucleus
DNA damage and repair
Growth arrest or apoptosis
DNA synthesis
Metastasis
ECM degradation
Tumour migration and invasion
Motility factors
Angiogenic factors
Growth factors
Endothelial cell proliferation tube formation
Growth effects
Proliferation
Differentiation
Angiogenesis blood vessel recruitment

Proposed model of action of ^{111}In-NLS-Nmab

Objectives

Hypothesis

^{111}In-NLS-Nmab (EGFR mAb) will be able to deliver lethal, DNA damaging doses of radiation specifically to the nucleus of trastuzumab resistant and EGFR-overexpressing BC cells while minimizing toxicity towards normal tissues.

Objectives

1. Development and characterization of ^{111}In-NLS-Nmab
2. Analyze receptor binding properties by using direct radioligand binding assays
3. Cell internalization and nuclear localization studies
4. Clonogenic assays to evaluate ^{111}In-NLS-Nmab effects

Breast Cancer Cell Lines

1. MDA-MB-468: High EGFR expression: Insensitive to trastuzumab
2. MDA-MB-231:
3. TrR1: Intermediate EGFR expression: Low sensitivity to trastuzumab
4. TrR2:
5. MCF-7:

(TrR2, MCF-7) Low EGFR expression: Insensitive to trastuzumab

EGFR expression (receptors/cell):
High (0.5-1.0 x 10^6), Intermediate (0.2-0.7 x 10^5) and Low (<0.2 x 10^5)

Results

Construction and Characterization of ^{111}In-NLS-Nmab

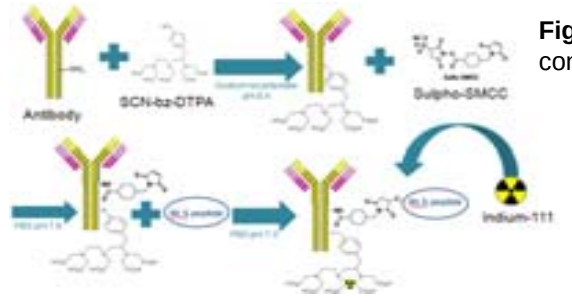
Antibody
SCN-bz-DTPA
Sulpho-SMCC
Indium-111

Fig.1. Diagram showing construction of ^{111}In-NLS-Nmab

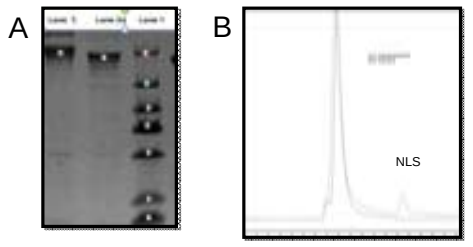
A
B
NLS

Fig.2. (A) SDS PAGE shows the conjugation of Nmab with NLS (8.5$\pm$0.20NLS per antibody) indicated by the band shift. Lane 2a shows the unconjugated antibody and lane 3 shows the NLS-Nmab (B) HPLC chromatogram of Nmab (Black) and ^{111}In-NLS-Nmab (Green). This indicates that conjugate is pure and no aggregates were formed during conjugation.

Radioligand Binding Assay

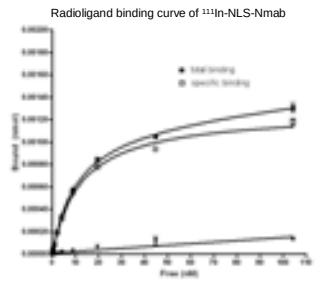
Radioligand binding curve of 111In-NLS-Nmab

Fig.3. Characterization of the binding of ^{111}In-Nmab to EGFR on MDA-MB-468 cells by radioligand saturation binding assays. The dissociation constant for binding of ^{111}In-NLS-Nmab to MDA-MB-468 cells was 9.8$\pm$0.862 nM and Bmax was 0.00121nmol.

Internalization Assay

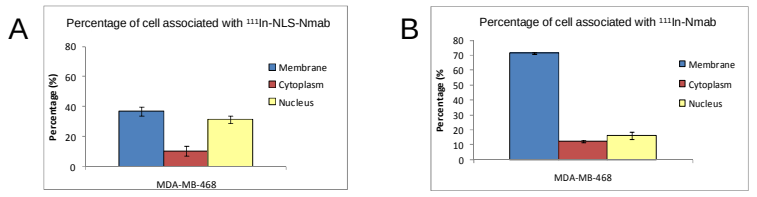
A
Percentage of cell associated with 111In-NLS-Nmab
B
Percentage of cell associated with 111In-Nmab
Membrane
Cytoplasm
Nucleus
Percentage (%)
MDA-MB-468

Fig.4. Internalization assay with respect to the radioactivity associated to the cells. (A) Internalization of ^{111}In-NLS-Nmab by MDA-MB-468 BC cells. (B) Internalization of ^{111}In-Nmab by MDA-MB-468 cells.

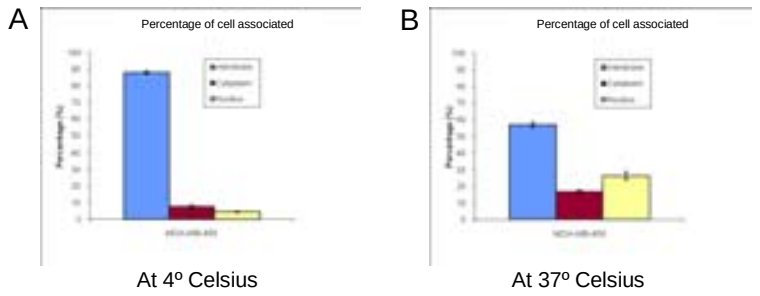
A
Percentage of cell associated
B
Percentage of cell associated
At 4º Celsius
At 37º Celsius

Fig.5. Internalization of ^{111}In-NLS-Nmab at 4ºC and 37ºC in MDA-MBA-468 cells with respect to the radioactivity associated with the cells (A) shows internalization at 4ºC and (B) shows internalization at 37ºC .

Internalization Assay

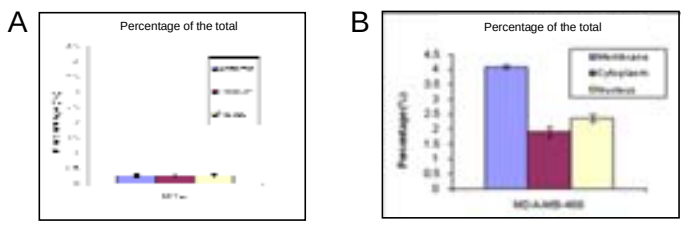
A
Percentage of the total
B
Percentage of the total

Fig.6. Internalization of ^{111}In-NLS-Nmab in (A) MCF-7 BC cell line and (B) MDA-MB-468 with respect to the total amount of radioactivity added to incubation media.

Clonogenic Assay

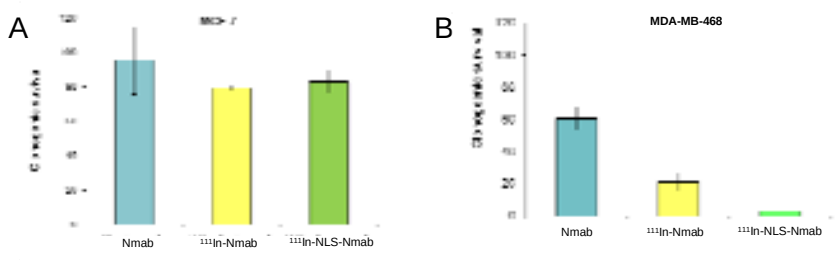
A
B
MDA-MB-468
Nmab
111In-Nmab
111In-NLS-Nmab

Fig. 7. Clonogenic survival of cells treated for 3 days with Nmab or ^{111}In-Nmab or ^{111}In-NLS-Nmab (A) MCF-7 cells (B) MDA-MB-468 cells.

Future Studies

1 *In vitro studies*

1. Do further clonogenics on Herceptin resistant cell lines such as MDA-MB-231, TrR1, TrR2
2. Gamma H2AX assay to assess DNA damage
3. Confocal microscopy studies to visualize nuclear importation

2 *In vivo* studies with ^{111}In-NLS-Nmab in Breast Cancer Xenografts

1. Biodistribution studies in athymic mice to evaluate tumour and normal tissue localization
2. PET imaging studies to measure tumor response

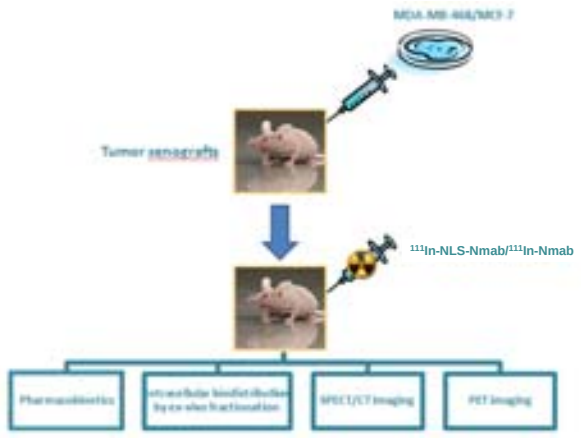
Tumor xenografts
111In-NLS-Nmab/111In-Nmab

Conclusions

- The binding of ^{111}In-NLS-Nmab to EGFR on MDA-MB-468 cells was conserved and the NLS promoted higher nuclear uptake compared to ^{111}In-Nmab without NLS.
- NLS-peptides routed ^{111}In-Nmab into the nucleus of EGFR-positive human BC cells, rendering the radiopharmaceutical lethal through the emission of nanometer range Auger electrons.
- The greater cytotoxic potency of ^{111}In-NLS-Nmab compared to Nmab *in vitro* and its ability to discriminate between cells with overexpressed EGFR (e.g. MDA-MB-468) compared to normal levels of EGFR (e.g. MCF-7) suggests that it could be an effective targeted radiotherapeutic agent for EGFR-positive and trastuzumab-resistant BC in humans.

References and Acknowledgements

1. Harris AL, Nicholson S, Sainsbury R, Wright C, Farndon J. Epidermal growth factor receptor and other oncogenes as prognostic markers. *J Natl Cancer Inst Monogr*. 1992(11):181-7.
2. Sainsbury JRC, Farndon JR, Needham GK. Epidermal-growth-factor receptor status as predictor of early recurrence of and death from breast cancer. *Lancet*. 1987;1(8547):1398-402.
3. Spicer J. Technology evaluation: Nimotuzumab, the center of molecular Immunology/YM BioSciences/Oncoscience. *Curr Opin Mol Ther*. 2005;7(2):182-91.
4. Snyder LC, Astsaturov I, Weiner LM. Overview of monoclonal antibodies and small molecules targeting the epidermal growth factor receptor pathway in colorectal cancer. *Clin Colorectal Cancer*. 2005;5(SUPPL. 2)
5. Allan DGP. Nimotuzumab: Evidence of clinical benefit without rash. *Oncologist*. 2005;10(9):760-1.

This research was supported by grants from the Canadian Breast Cancer Research Alliance (CBCRA). AF is a recipient of MDS Nordion Award and Ben–Cohen Bursary.