ABSTRACT # 41363

Safety and Efficacy of CYT387, a JAK1 and JAK2 Inhibitor for the Treatment of Myelofibrosis

A. Pardanani[1], J. Gotlib[2], V. Gupta[3], A. W. Roberts[4], M. Wadleigh[5], S. Sirhan[6], M. R. Litzow[1], W. J. Hogan[1], K. Begna[1], G. Smith[7], L.M. Bavisotto[8], M. Kowalski[7], and A. Tefferi[1]

[1]Mayo Clinic (Rochester); [2]Stanford Cancer Center; [3]Princess Margaret Hospital; [4]Royal Melbourne Hospital; [5]Dana Farber Cancer Institute; [6]Jewish General Hospital; [7]YM BioSciences Inc.; [8]Porta Clinica LLC

BACKGROUND

Study

- CYT387 is a potent JAK1 and JAK2 inhibitor. The maximum tolerated dose (MTD) and preliminary safety and efficacy of once-daily dosing of CYT387 were determined in a single center Phase I/II study. Previously reported results from the first 60 subjects enrolled in this study determined a MTD of 300 mg QD and demonstrated improvements in splenomegaly and constitutional symptoms as well as in red blood cell (RBC) transfusion requirements. The study was expanded into a multicenter study with the addition of a twice–daily dosing arm. The accrual of 166 subjects at six study sites is complete. The preliminary safety and efficacy results from this expanded multicenter study are presented.

CYT387

- CYT387 is a low nanomolar, ATP-competitive inhibitor of JAK2 and JAK1. It is broadly selective for JAK1 and JAK2 over other kinase enzymes, including the closely related JAK3 and TYK2.
- CYT387 displays potent *in vitro* inhibitory activity against cells dependent on JAK2, including the JAK2V617F mutant which is associated with the etiology of Polycythemia Vera (PV) and the other Myeloproliferative Neoplasms (MPNs). CYT387 has also been shown to potently inhibit JAK2 signaling and the formation of V617F mutant myeloid colonies in a dose-responsive manner. In *ex vivo* assays of human erythroid cells from PV patients, and in a transgenic mouse model of the MPNs, CYT387 has been demonstrated to have marked disease-modifying properties.
- The growing clinical validation of JAK2 inhibition as a treatment modality in these disorders, and this favorable preclinical data for CYT387, support the clinical development of CYT387 as a treatment modality in the MPNs.

STUDY DESIGN

A Phase I/II open-label, non-randomized, dose-escalation study. Conducted in three phases:

1. Dose-escalation phase (Part 1), determined the safety and tolerability of CYT387.
 - Dose-limiting toxicities (DLTs) (2 of 6 subjects at 400 mg QD) were asymptomatic, reversible Gr 3 hyperlipasemia (n=1), and Gr 3 headache (n=1).
 - Identified the MTD for CYT387 as 300 mg QD.
2. Dose-confirmation phase (Part 2), a cohort expansion at or below the MTD of CYT387. The dosages determined for Part 2 were 150 mg QD and 300 mg QD.
3. Dose-expansion phase (Part 3), carried out at multiple study sites to identify a therapeutic dose for CYT387. The dosages determined for Part 3 were 150 mg QD, 300 mg QD and 150 mg BID.

- CYT387 was orally self-administered beginning on Day 1 of the study, and thereafter at approximately the same time(s) each day of a 28-day cycle.
- The Core study consists of nine treatment cycles. Evaluations were performed weekly during the first cycle, every two weeks for cycles two and three, and at the end of each subsequent cycle.
- Patients who achieve at least stable disease or better (per IWG-MRT criteria) and tolerate the drug well may continue to receive CYT387 beyond the planned nine cycles in an Extension phase of the study.

Study Objectives

The primary objectives of this study are:

- To determine the safety and tolerability, DLTs and MTD of orally-administered CYT387 in patients with Primary Myelofibrosis (PMF) or post Essential Thrombocythemia/Polycythemia Vera (post-ET/PV) Myelofibrosis (MF).
- To obtain preliminary information on the effectiveness of orally-administered CYT387 in patients with PMF or post-ET/PV MF, as measured according to IWG-MRT criteria.

Key Entry Criteria

- Diagnosis of PMF or post-ET/PV MF as per revised World Health Organization (WHO) criteria.
- High-risk or Intermediate-2 risk MF (as defined by the International Prognostic Scoring System [IPSS]); or Intermediate-I risk MF (IPSS) associated with symptomatic splenomegaly/hepatomegaly and/or unresponsive to available therapy.
- Must have evidence of acceptable organ function within seven days of initiating study drug as evidenced by the following:
 - SGOT (AST) or SGPT (ALT) ≤ 2.5 x upper limit of normal (ULN) (or ≤ 5 x ULN if in the investigator's opinion the elevation is due to extramedullary hematopoiesis)
 - Bilirubin ≤ 2.0 x ULN or direct bilirubin < 1.0
 - Serum creatinine ≤ 2.5 x ULN
 - Absolute neutrophil count ≥ 500/μL
 - Platelet count ≥ 50,000/μL
- No chemotherapy, immunomodulatory drug therapy, immunosuppressive therapy or corticosteroids >10 mg/day prednisone or equivalent, or growth factor treatment within 14 days prior to initiation of study drug.

Current Study Status

Dose Escalation Phase (n=21) → Dose Confirmation Phase (n=39) → Dose Expansion Phase (n=106) → Extension Phase (n=104)

- Enrollment is complete. Data collection and analysis are ongoing.
- Extension Phase: 97% of patients who completed the Core study entered the Extension phase.

RESULTS

Subject Disposition

- Median follow–up (range) for Core and Extension: 10.4 months (0.8–25.6)*
- Median follow–up (range) for Core: 9.0 months (0.8-9.0)*
- Study discontinuation during Core: n=32 (19%); 81% retention rate
- Reasons for discontinuation during Core (n=32):
 - Unrelated comorbid conditions (n=11)
 - Subject withdrew consent (n=5)
 - Possibly or probably related adverse event (n=5)
 - Neuropathy (n=2), cough (n=1), transaminitis (n=1), thrombocytopenia (n=1)
 - Disease progression (n=4)
 - Investigator's decision (n=4)
 - Stem cell transplant (n=2)
 - Intercurrent illness (n=1)
- Deaths during Core: n=12 (7%)
- Study discontinuation during Extension: n=16 (21%); 79% retention rate

* ongoing

Subject Baseline Characteristics

Characteristic	Value
Number of Subjects	166
Age (years)	
Median	67
Range	34–89
Sex	
Male	58%
Female	42%
Myelofibrosis	
Primary	65%
Post-polycythemia vera	22%
Post-essential thrombocythemia	14%
JAK2V617F Positive	76%
DIPSS-Plus Category*	
Int-1	11%
Int-2	61%
High	28%

Characteristic	Value
Prior Therapy	
JAK2 inhibitor (INCB018424, TG101348, Unspecified)	12%
IMiDs (Pomalidomide, Thalidomide, Revlimid)	9%
Palpable Splenomegaly > 10 cm	80%
Spleen Size (cm) (Spleen Evaluable; n=142)	
Mean (STD)	18.2 (6.5)
Median	17.0
RBC Transfusion Dependent	44%
Median Hemoglobin (g/dL)	
All subjects (n=166)	9.5
Transfusion independent subjects (n=93)	10.4
Transfusion dependent subjects (n=73)	8.7
Platelet Count (10^9/L)	
Median	187.5

*Based on available data; includes unfavorable karyotype, platelet count and transfusion dependent status as additional prognostic factors.

SAFETY

Related Non-Hematologic Adverse Events

Adverse Event (n=166) Incidence ≥ 10%	All Grades	Grade 3	Grade 4
Dizziness[1]	20%	0	0
Neuropathy peripheral	20%	0	0
Diarrhea	18%	0	0
Nausea	17%	0	<1%
Headache	12%	1%	0

Adverse Event (n=166)	All Grades	Grade 3	Grade 4
First Dose Effect[2]	20%	0	0
Dizziness	11%	0	0
Flushing	5%	0	0
Hypotension	6%	0	0

1 Includes First Dose Effect
2 Only events occurring on first dose date

At least possibly related to study drug
Common Terminology Criteria for Adverse Events v3.0

Related Non-Hematologic Laboratory Adverse Events

Adverse Event (n=166) Incidence ≥ 5%	All Grades	Grade 3	Grade 4
ALT increased	8%	2%	0
Lipase increased	8%	4%	0
AST increased	7%	1%	<1%
Amylase increased	6%	0	0
Bilirubin increased	6%	0	0
Creatinine increased	6%	0	0

At least possibly related to study drug
Common Terminology Criteria for Adverse Events v3.0

Related Hematologic Adverse Events

Adverse Event (n=166)	All Grades	Grade 3	Grade 4
Thrombocytopenia	33%	11%	6%
Baseline platelets > 100 x 10^9/L	26%	6%	2%
Baseline platelets > 200 x 10^9/L	12%	4%	0
Neutropenia	6%	1%	2%
Anemia	4%	1%	0
Leukopenia	2%	<1%	<1%
Leukocytosis	1%	<1%	0

At least possibly related to study drug
Common Terminology Criteria for Adverse Events v3.0

RESPONSE

Spleen Response

Response by Dose	150 mg QD (n=52)	300 mg QD (n=60)	150 mg BID (n=42)	Total[1] (n=166)
Spleen evaluable (n)	47	51	33	142
Median time on study* (days)	252	225	144	225
Spleen response* (IWG-MRT)	30%	33%	27%	31%
≥50% decrease in palpable spleen length at six months	28%	33%	39%	33%
Median spleen decrease at six months	-35%	-35%	-39%	-35%

Response by Diagnosis	Primary MF (n=106)	Post-PV MF (n=36)	Post-ET MF (n=24)
Spleen evaluable (n)	88	34	20
Spleen response* (IWG-MRT)	28%	38%	30%

Time to Response	Median	Min-Max
Time to IWG-MRT response (days)	15	6-260
Duration of response (days)	not yet reached	55-574*

* ongoing
1 Includes 100 mg QD (n=3), 200 mg QD (n=3), and 400 mg QD (n=6) doses

Maximal Change in Palpable Spleen Size* (Core Study; n=142)

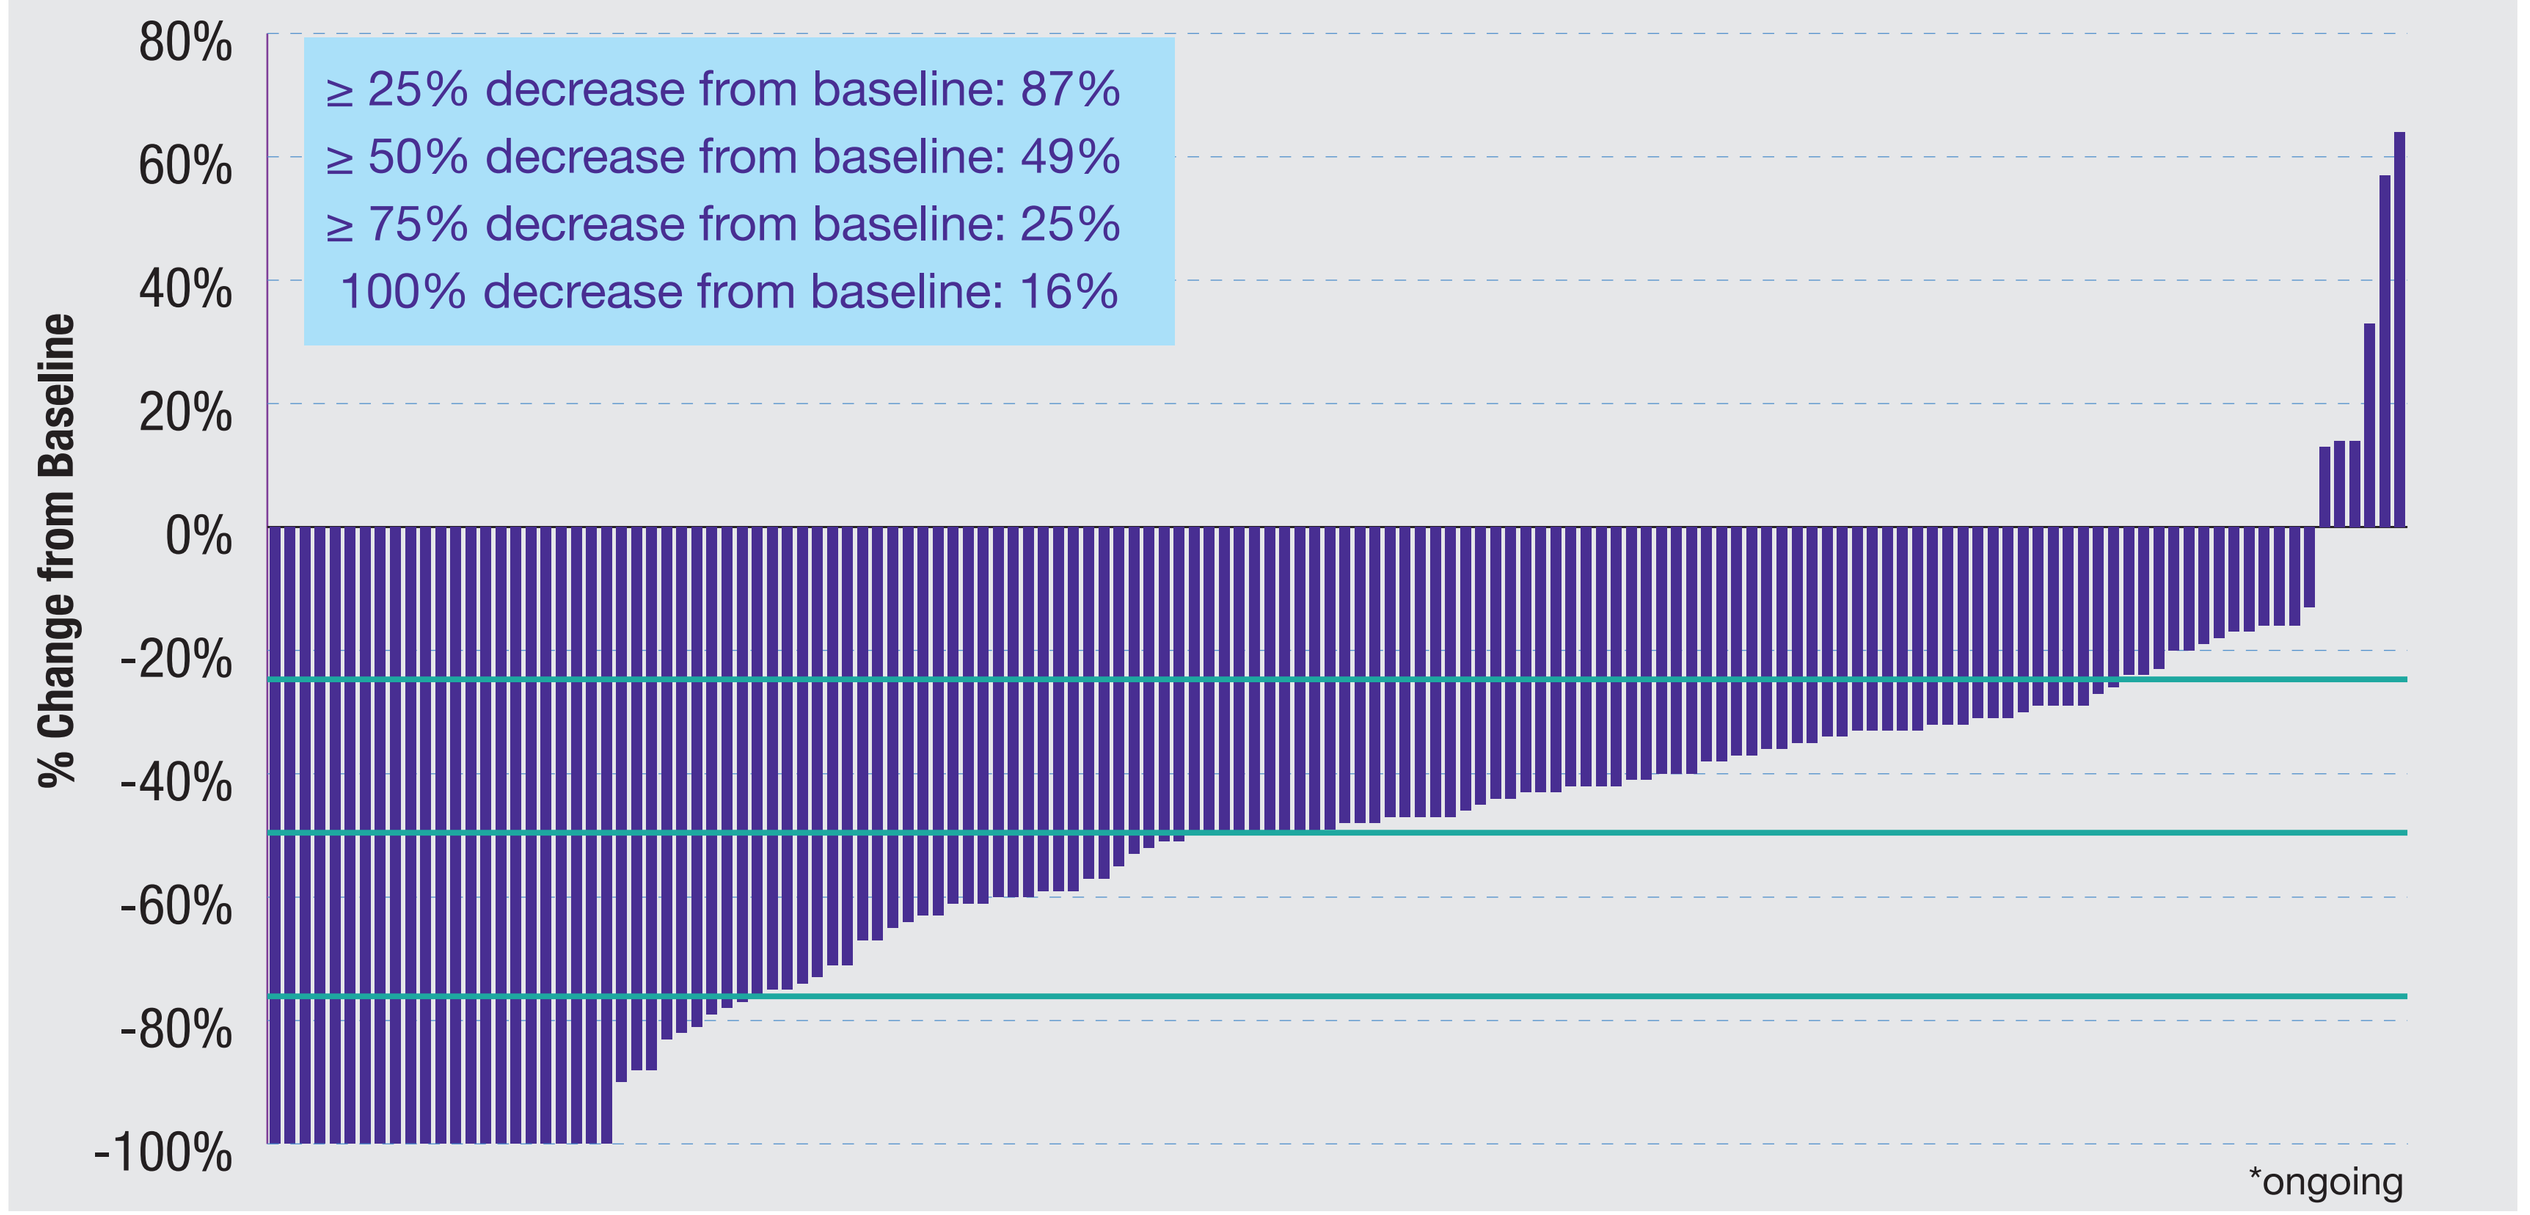
≥ 25% decrease from baseline: 87%
≥ 50% decrease from baseline: 49%
≥ 75% decrease from baseline: 25%
100% decrease from baseline: 16%
% Change from Baseline
80%
60%
40%
20%
0%
-20%
-40%
-60%
-80%
-100%
*ongoing

Spleen Response: MRI vs. Palpation

	MRI[1] (n=11)	Palpation (n=11)
Response at three months	64%[2]	45%[3]
Median decrease from baseline at three months	-41%	-45%

- Median spleen volume at baseline: 3667 cm^3

Spleen Response: Change from Baseline at Three Months

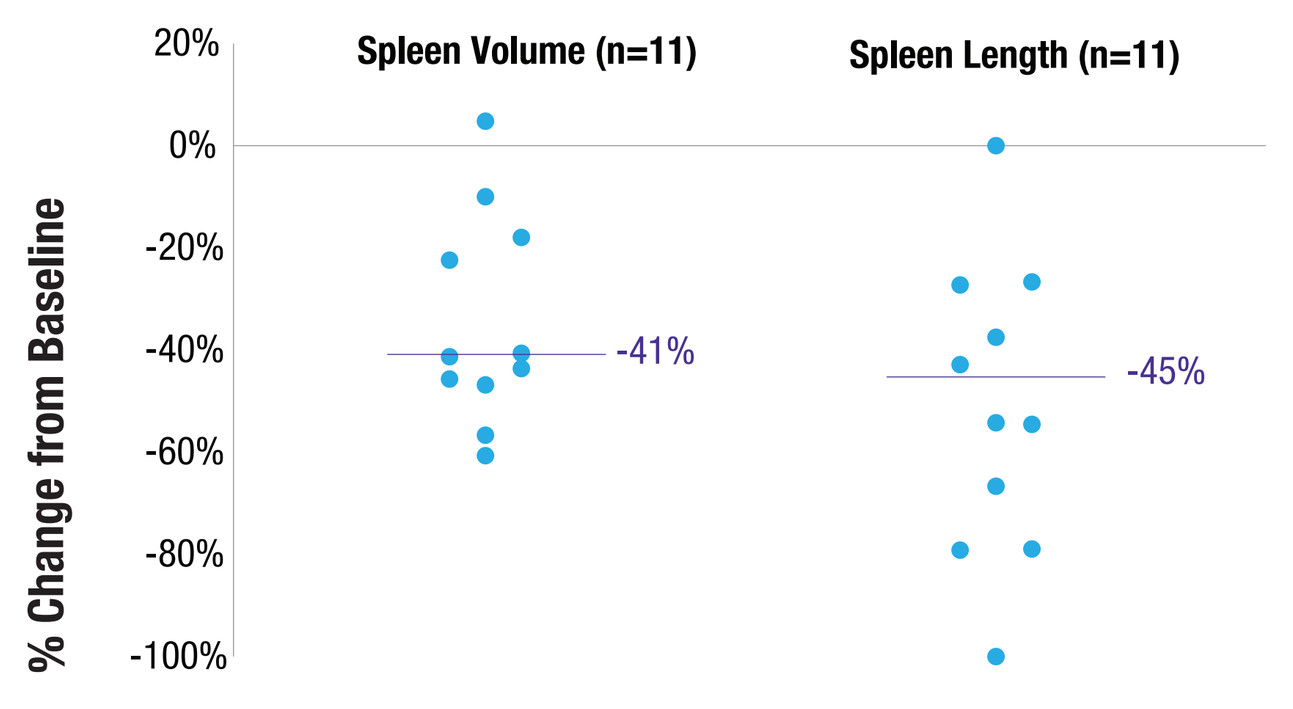
Spleen Volume (n=11)
Spleen Length (n=11)
% Change from Baseline
20%
0%
-20%
-40%
-60%
-80%
-100%
-41%
-45%

1 Includes 1 subject with CT assessment
2 Spleen response defined as a 35% reduction in spleen volume from baseline
3 Spleen response defined as a 50% reduction in spleen length from baseline

Anemia Response

Transfusion Independence Response

Response by Dose	150 mg QD (n=52)	300 mg QD (n=60)	150 mg BID (n=42)	Total[1] (n=166)
Transfusion dependent at baseline (evaluable; n)	25	26	14	68
Median time on study (days)	251	245	141	250
Transfusion independence rate (12 weeks)*	48%	65%	43%[2]	54%
Transfusion independence rate (12 weeks & Hgb≥8g/dL)*	40%	62%	29%[2]	46%

1 Includes 100 mg QD (n=3), 200 mg QD (n=3), and 400 mg QD (n=6) doses
2 Not statistically significant vs. 300 mg QD
*ongoing

- >25% of subjects not receiving transfusions while on study experienced at least a 1 g/dL increase in Hgb for ≥ 8 weeks.

Time to Response	Median	Min-Max
Time to confirmed response (12 wks & Hgb≥8 g/dL) (days)	84	84-293
Duration of transfusion-free period (12 wks & Hgb≥8 g/dL) (days)	not yet reached	82-506*

*ongoing

Maximal Duration of Transfusion-Free Period*

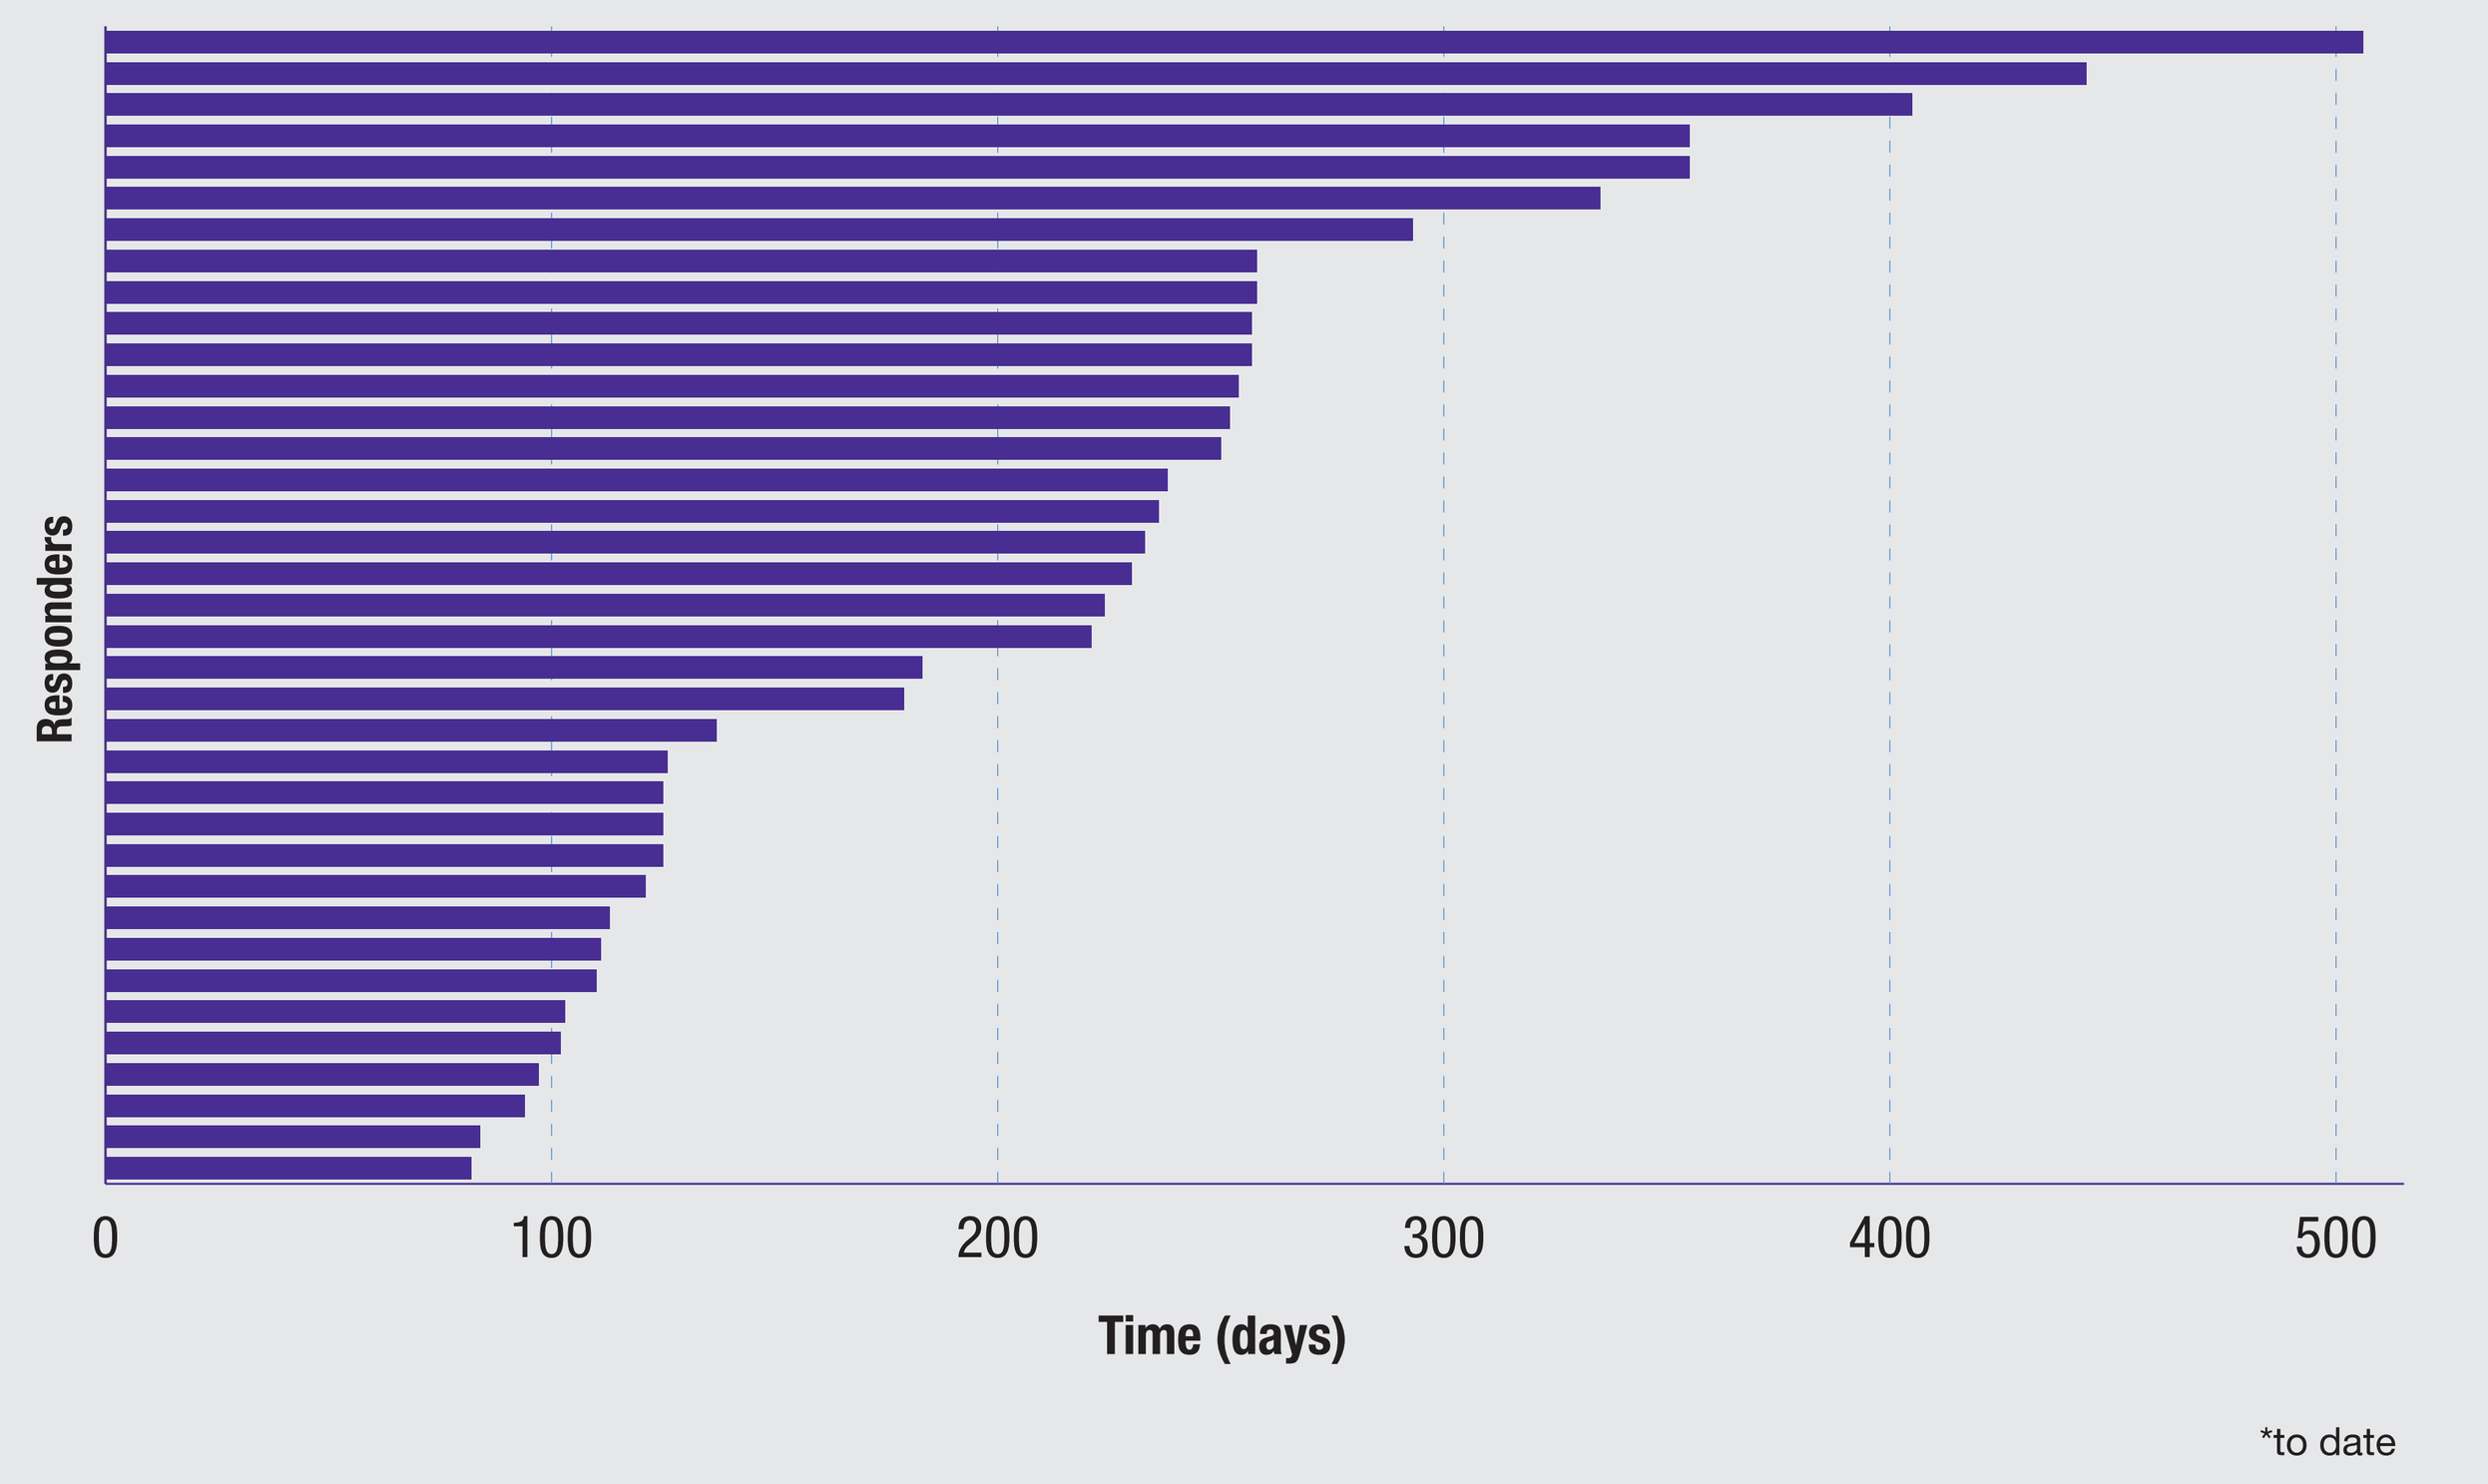
Responders
0
100
200
300
400
500
Time (days)
*to date

Constitutional Symptoms Response

Constitutional Symptoms Response at Six Months

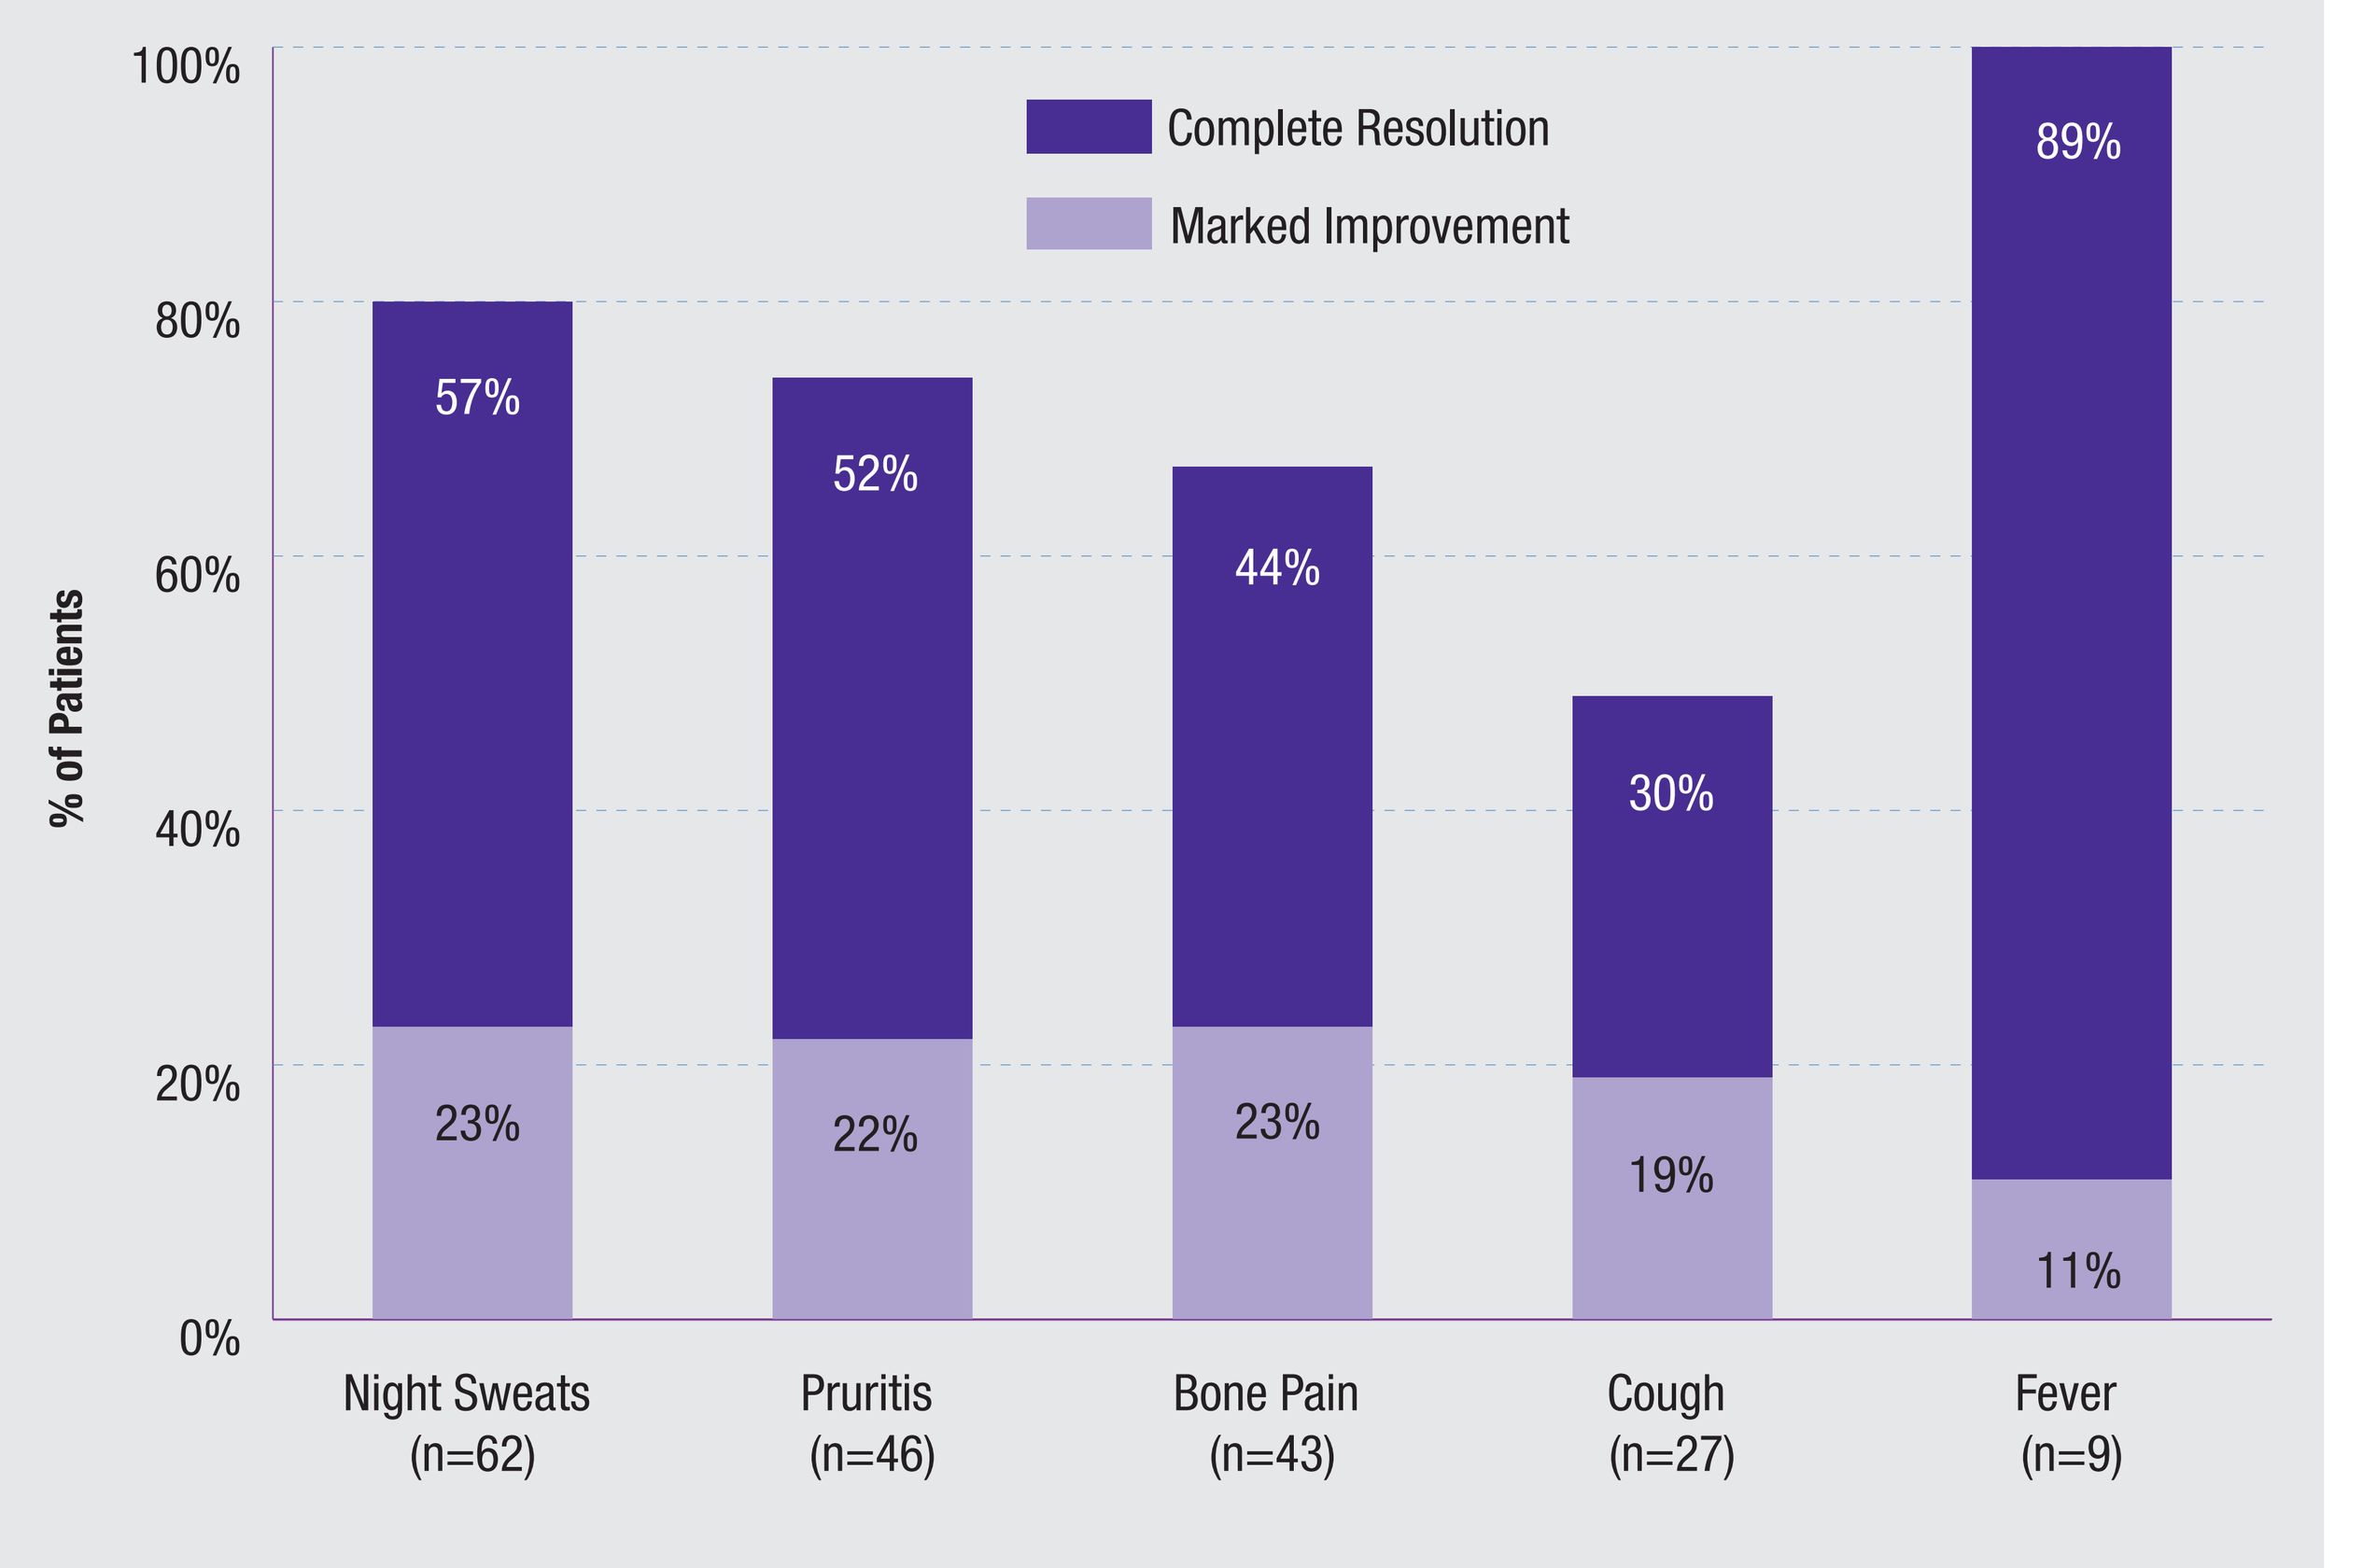
Complete Resolution
Marked Improvement
% of Patients
100%
80%
60%
40%
20%
0%
57%
23%
52%
22%
44%
23%
30%
19%
89%
11%
Night Sweats (n=62)
Pruritis (n=46)
Bone Pain (n=43)
Cough (n=27)
Fever (n=9)

CONCLUSIONS

- CYT387 is well tolerated in myelofibrosis patients for dosing periods up to and exceeding two years.
 - Reported adverse effects include thrombocytopenia; transient, mild dizziness; mild peripheral neuropathy; and abnormalities in liver/pancreas-related laboratory tests
 - Treatment-emergent anemia and neutropenia are rare
- CYT387 treatment results in significant, durable responses in anemia, splenomegaly and constitutional symptoms at 150 mg QD, 300 mg QD and 150 mg BID dose levels.
 - Therapeutic benefit and safety established in a population with multiple risk factors, including anemia and thrombocytopenia
 - CYT387 anemia benefit appears unique among the current class of JAK1 and JAK2 inhibitors
 - Clinically relevant maintenance of transfusion independence period
 - MRI performed in a subset of subjects confirms the meaningful improvement in splenomegaly measured by palpation
 - Complete resolution or marked improvement of common constitutional symptoms is achieved in the majority of subjects
- Although additional assessments and analyses are ongoing, 300 mg QD appears to be a safe and effective dosing regimen that warrants further clinical development.